P.378

Efficacy and Safety of Nusinersen in Children With Later-Onset Spinal Muscular Atrophy (SMA): End of Study Results From the Phase 3 CHERISH Study

Mercuri E,[1] Finkel RS,[2] Kirschner J,[3] Chiriboga CA,[4] Kuntz N,[5] Sun P,[6] Gheuens S,[6] Bennett CF,[7] Schneider E,[7] Farwell W,[6] on behalf of the CHERISH Study Group

[1]Università Cattolica del Sacro Cuore, Roma, Italy; [2]Nemours Children's Health System, Orlando, FL, USA; [3]Universtatsklinikum Freiburg, Freiburg, Germany; [4]Columbia University, New York, NY, USA; [5]Ann & Robert H. Lurie Children's Hospital of Chicago, Chicago, IL, USA; [6]Biogen, Cambridge, MA, USA; [7]Ionis Pharmaceuticals, Inc., Carlsbad, CA, USA

Presenting author e-mail: eumercuri@gmail.com

Conclusions

- **In the CHERISH study, nusinersen demonstrated significant and clinically meaningful improvements in motor function vs. sham procedure, as assessed by the HFMSE from baseline to Month 15.**
 - **Improvements for nusinersen vs. sham procedure also were observed in the number of new World Health Organization motor milestones achieved per child and in upper limb function.**
- **Nusinersen demonstrated a favorable safety profile, and no children discontinued treatment due to AEs.**
 - **The majority of AEs were considered to be related to SMA, common events in the general population or events related to the lumbar puncture procedure.**
- **Children from CHERISH have been transitioned into the SHINE (NCT02594124) open-label extension study.**

Introduction

- Nusinersen is an antisense oligonucleotide approved for the treatment of spinal muscular atrophy (SMA).[1,2]
- Nusinersen has demonstrated significant and clinically meaningful efficacy on the achievement of motor milestones and measures of motor function, as well as favourable safety across multiple SMA populations, and significantly greater event-free survival vs. sham procedure in infants with infantile-onset SMA (most likely to develop SMA Type I).[3-5]

Objectives

- CHERISH (NCT02292537) was a Phase 3, multicentre, randomised, double-blind, sham procedure–controlled study to assess the efficacy and safety of nusinersen in children with later-onset SMA (most likely to develop SMA Type II or III).

Methods

- Children with symptomatic SMA 2–12 years of age were randomised 2:1 (stratified based on screening age <6 vs. ≥6 years) to receive 4 doses of intrathecal nusinersen (12 mg non-scaled) or sham procedure over 9 months during this 15-month study.
- Key inclusion criteria included confirmed 5q SMA and onset of SMA clinical symptoms at ≥6 months of age.
- The primary endpoint was change from baseline in Hammersmith Functional Motor Scale Expanded (HFMSE) score at Month 15.
 - An interim analysis was pre-specified when all children had completed their 6-month assessment and ≥39 children had completed their 15-month assessment.

Results

- Baseline demographics were generally similar between groups, with slight differences in age, sex and race (Table 1).
- At the pre-specified interim analysis, there was a significant treatment difference of 5.9 points in mean HFMSE score changes from baseline to Month 15 with a 4.0-point mean improvement observed with nusinersen vs. a mean decline of 1.9 points with sham procedure (*P*=.0000002; Figure 1A).
- In the end of study analysis, the treatment difference in change from baseline to Month 15 in mean HFMSE score also was highly clinically and statistically significant (4.9 points: nusinersen, 3.9-point improvement; sham procedure, 1.0-point decline; nominal *P*=.0000001; Figure 1B–C).
- Treatment-emergent adverse events (AEs) are listed in Table 3.
 - There was no evidence of adverse effects on platelet counts, renal function or hepatic enzymes.

Table 1. Baseline characteristics

Characteristic	Sham procedure n=42	Nusinersen n=84
Female, n (%)	21 (50)	46 (55)
Median (range) age at screening, y	3.0 (2–7)	4.0 (2–9)
Median (range) age at symptom onset, mo	11.0 (6–20)	10.0 (6–20)
Median (range) age at SMA diagnosis, mo	18.0 (0–46)	18.0 (0–48)
Median (range) disease duration, mo	30.2 (10–80)	39.3 (8–94)
Children who have ever achieved motor milestone, n (%)		
Sat without support	42 (100)	84 (100)
Walked with support	14 (33)	20 (24)
Stood without support	12 (29)	11 (13)
Walked ≥15 ft independently	0	0
Children using a wheelchair, n (%)	29 (69)	64 (76)
SMN2 gene copies, n (%)		
2	4 (10)	6 (7)
3	37 (88)	74 (88)
4	1 (2)	2 (2)
Unknown	0	2 (2)
Mean (SD) HFMSE total score[a]	19.9 (7.2)	22.4 (8.3)
Mean (SD) WHO total score[a,b]	1.5 (1.0)	1.4 (1.0)
Mean (SD) RULM total score[a,c]	18.4 (5.7)	19.5 (6.2)

HFMSE = Hammersmith Functional Motor Scale Expanded; RULM = Revised Upper Limb Module; SMA = spinal muscular atrophy; SMN = survival of motor neuron; WHO = World Health Organization
[a]Baseline was defined as the last non-missing value before the first dose of nusinersen or sham procedure
[b]If the baseline value as defined above was missing, then baseline was imputed as the median of the non-missing values of the stratum to which the child belongs: age <6 vs. ≥6 years
[c]One child had a missing value and this was imputed as the median baseline value of the child across all the multiply imputed datasets

Table 2. Secondary endpoints: WHO motor milestones at Month 15

Endpoint	Sham procedure n=42	Nusinersen n=84	Treatment difference
LSM (95% CI) no. of new motor milestones achieved per child[a]	–0.2 (–0.4 to 0.0)	0.2 (0.1 to 0.3)	Nominal *P*=.0001[b]
% (95% CI) of children achieving standing alone[c]	2.9 (0.07 to 15.3)	1.5 (0.04 to 8.2)	Nominal *P*>.9999[d]
% (95% CI) of children achieving walking with assistance[c]	0 (0 to 10.3)	1.5 (0.04 to 8.2)	Nominal *P*>.9999[e]

LSM = least squares mean; WHO = World Health Organization
[a]Based on analysis of covariance with treatment as a fixed effect and adjustment for each child's age at screening and number of motor milestones at baseline
[b]LSM difference: 0.4 (95% CI, 0.2 to 0.7)
[c]Proportion of responders based on exact CI. Difference in proportions based on exact unconditional CI. *P* value based on Fisher's exact test
[d]Difference in proportions: –1.4 (95% CI, –21.8 to 19.3)
[e]Difference in proportions: 1.5 (95% CI, –19.1 to 22.1)

Figure 1. Mean change from baseline in HFMSE score at: **(A)** pre-specified interim analysis (primary endpoint); **(B)** end of study; and **(C)** over time at end of study

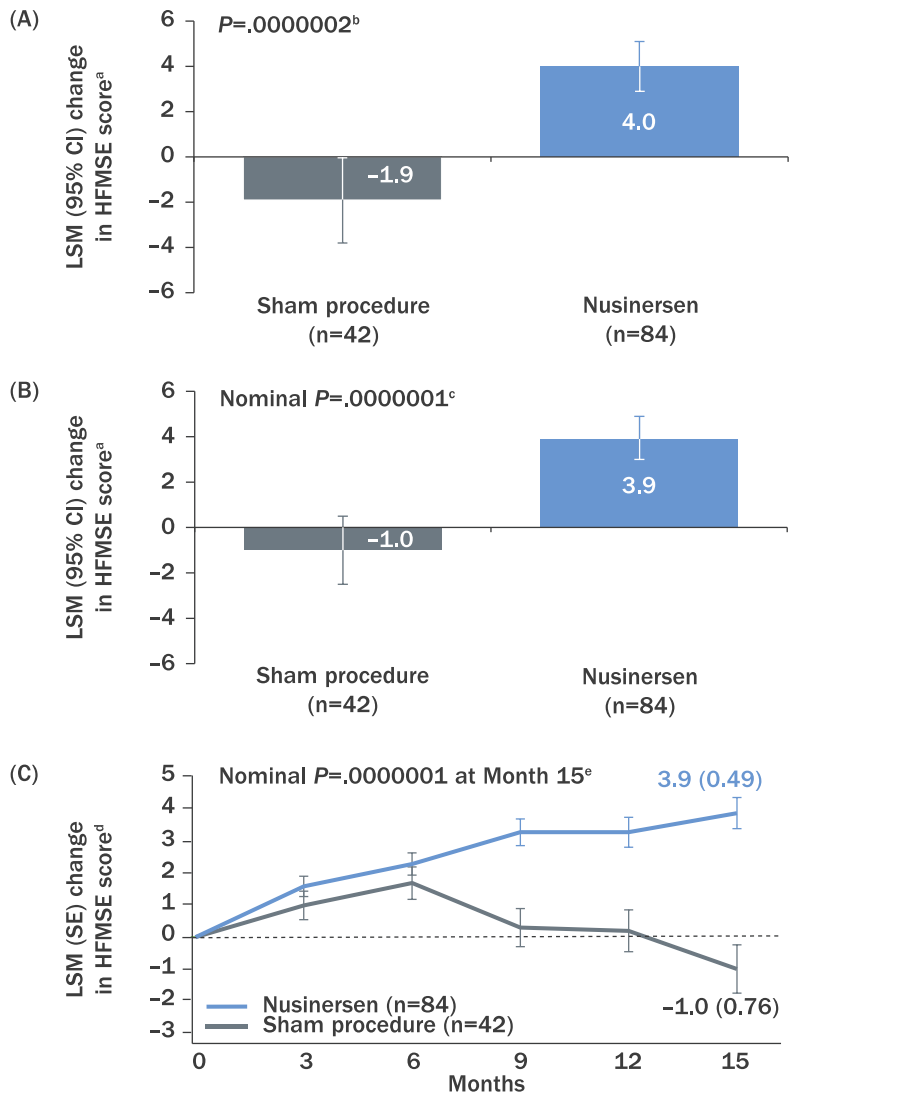

(A)
P=.0000002[b]
LSM (95% CI) change in HFMSE score[a]
–1.9
4.0
Sham procedure (n=42)
Nusinersen (n=84)
(B)
Nominal *P*=.0000001[c]
LSM (95% CI) change in HFMSE score[a]
–1.0
3.9
Sham procedure (n=42)
Nusinersen (n=84)
(C)
Nominal *P*=.0000001 at Month 15[e]
LSM (SE) change in HFMSE score[d]
3.9 (0.49)
–1.0 (0.76)
Nusinersen (n=84)
Sham procedure (n=42)
Months

HFMSE = Hammersmith Functional Motor Scale Expanded; LSM = least squares mean
[a]From baseline to Month 15. Based on multiply imputed (MI) data. From MI procedure, based on analysis of covariance (ANCOVA) with treatment as a fixed effect and adjustment for each child's age at screening and HFMSE score at baseline. These estimates were constructed from fitting the ANCOVA model to each of the imputed datasets. Interim analysis: observed: sham procedure, n=19; nusinersen, n=35; imputed: sham procedure, n=23; nusinersen, n=49. Final analysis: observed: sham procedure, n=34; nusinersen, n=66; imputed: sham procedure, n=8; nusinersen, n=18
[b]LSM difference: 5.9 (95% CI, 3.7 to 8.1)
[c]LSM difference: 4.9 (95% CI, 3.1 to 6.7)
[d]Change from baseline to each visit was analysed using an ANCOVA model and MI. LSM and LSM differences for treatment comparison based on MI procedure with ANCOVA fitted to each time point with treatment as a fixed effect and adjustment for each child's age at screening and HFMSE at baseline
[e]LSM difference: 4.9 (95% CI, 3.1 to 6.7)

Table 3. Overall summary of treatment-emergent AEs

AE, n (%)	Sham procedure n=42	Nusinersen n=84
Any AE	42 (100)	78 (93)
Moderate or severe AE	23 (55)	39 (46)
Severe AE	3 (7)	4 (5)
AE possibly related or related to study drug[a]	4 (10)	24 (29)
AE related to study drug[a]	0	1 (1)[b]
SAE	12 (29)	14 (17)
Most frequent AEs[c]		
Pyrexia	15 (36)	36 (43)
Upper respiratory tract infection	19 (45)	25 (30)
Headache	3 (7)	24 (29)
Vomiting	5 (12)	24 (29)
Back pain	0	21 (25)
Cough	9 (21)	21 (25)
Nasopharyngitis	15 (36)	20 (24)
Most frequent SAEs[d]		
Pneumonia	6 (14)	2 (2)
Influenza	2 (5)	0
Respiratory distress	2 (5)	2 (2)
Faecaloma	2 (5)	0
Dehydration	2 (5)	0
SAE related to study drug[a]	0	0
Discontinued treatment due to an AE	0	0
AEs observed at ≥5% higher frequency in nusinersen group 72 h after drug administration		
Back pain	0	19 (23)
Headache	1 (2)	22 (26)
Vomiting	1 (2)	11 (13)
Epistaxis	0	4 (5)

AE = adverse event; SAE = serious adverse event
[a]Investigator assessed relation to study drug
[b]One child had post-sedation nausea (procedural nausea) considered by the investigator to be related to study treatment
[c]Medical Dictionary for Regulatory Activities (MedDRA) Preferred Term (PT) in ≥20% of children in either treatment group
[d]MedDRA PT in ≥5% of children in either treatment group

Figure 2. Secondary endpoints at Month 15: **(A)** HFMSE responders; **(B)** proportion of children who achieved any new WHO motor milestone; and **(C)** change from baseline in RULM score

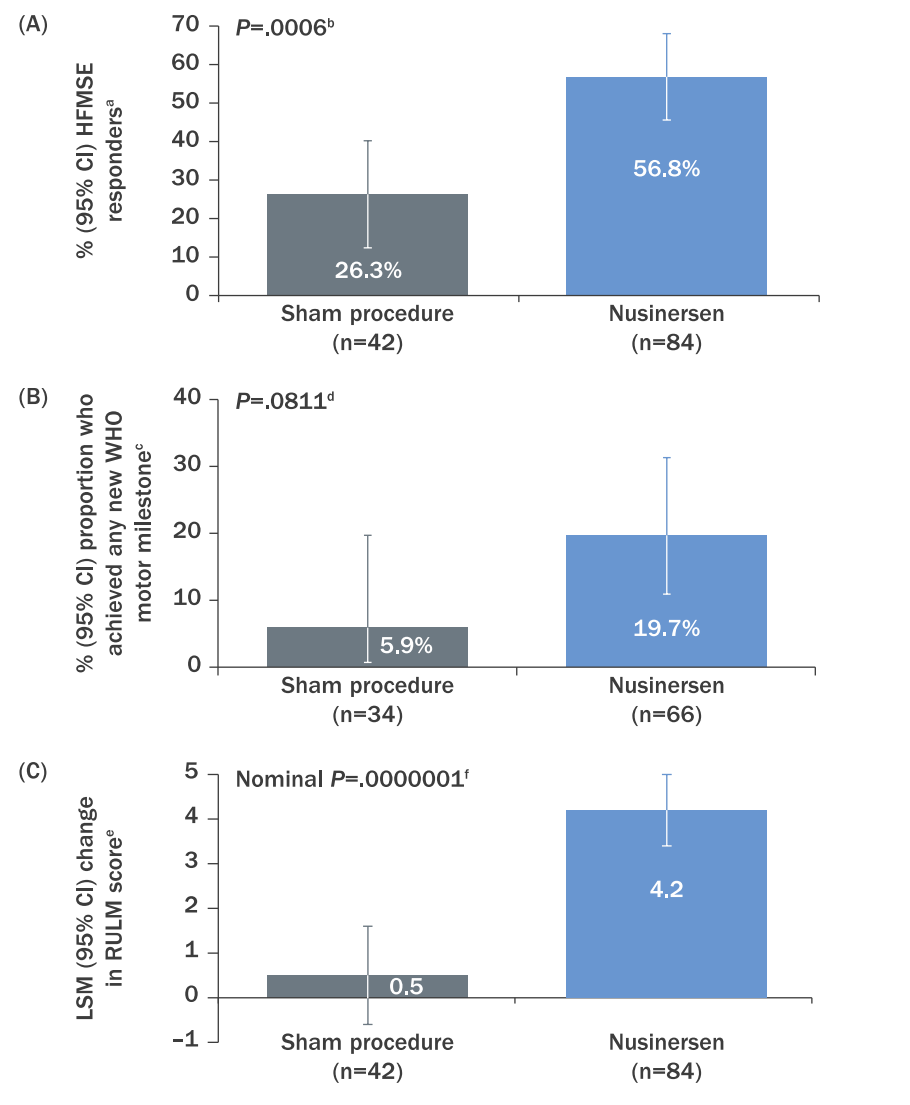

(A)
P=.0006[b]
% (95% CI) HFMSE responders[a]
26.3%
56.8%
Sham procedure (n=42)
Nusinersen (n=84)
(B)
P=.0811[d]
% (95% CI) proportion who achieved any new WHO motor milestone[c]
5.9%
19.7%
Sham procedure (n=34)
Nusinersen (n=66)
(C)
Nominal *P*=.0000001[f]
LSM (95% CI) change in RULM score[e]
0.5
4.2
Sham procedure (n=42)
Nusinersen (n=84)

HFMSE = Hammersmith Functional Motor Scale Expanded; LSM = least squares mean; RULM = Revised Upper Limb Module; WHO = World Health Organization
[a]HFMSE responder was defined as a child with an ≥3-point increase from baseline in HFMSE score at Month 15. If a child discontinued due to treatment failure or death then the child was classified as a non-responder irrespective of imputed value. Based on multiply imputed (MI) data. Estimates from the MI procedure were based on binomial proportions. Odds ratio based on logistic regression with treatment effect and adjustment for each child's baseline age and HFMSE score. Observed data: sham procedure, n=34; nusinersen, n=66
[b]Odds ratio: 5.59 (95% CI, 2.09 to 14.91)
[c]Children who maintained baseline WHO milestones at Month 15 and achieved ≥1 new milestone. Children who discontinued due to treatment failure or death before Month 15 were not included. Based on imputed data when there was missing data. Proportion of responders based on exact CI. Difference in proportions based on exact unconditional CI. *P* value based on Fisher's exact test
[d]Treatment difference: 13.8 (95% CI, –6.6 to 34.2)
[e]Based on MI data. From MI procedure, based on analysis of covariance (ANCOVA) with treatment as a fixed effect and adjustment for each child's age at screening and derived total score at baseline. Estimates were constructed from fitting the ANCOVA model to each of the imputed datasets. Observed data: sham procedure, n=34; nusinersen, n=66
[f]LSM difference: 3.7 (95% CI, 2.3 to 5.0)

References 1. Spinraza [prescribing information]. Cambridge, MA: Biogen; 2017. 2. European Medicines Agency. Spinraza 12 mg solution for injection [summary of product characteristics]. http://www.ema.europa.eu/docs/en_GB/document_library/EPAR_-_Product_Information/human/004312/WC500229704.pdf. Accessed August 30, 2017. 3. Finkel RS, *et al*. Primary efficacy and safety results from the phase 3 ENDEAR study of nusinersen in infants diagnosed with spinal muscular atrophy (SMA). Presented at: 43rd Annual Congress of the British Paediatric Neurology Association; January 11–13, 2017; Cambridge, UK. 4. Finkel RS, *et al*. *Lancet*. 2016;388(10063):3017-3026. 5. Bertini E, *et al*. Nusinersen in pre-symptomatic infants with spinal muscular atrophy (SMA): interim efficacy and safety results from the phase 2 NURTURE study. Presented at: 21st International Congress of the World Muscle Society; October 4–8, 2016; Granada, Spain.

Disclosures EM: advisory boards for AveXis, Biogen, Ionis Pharmaceuticals, Inc., Novartis and Roche; principal investigator for ongoing Ionis Pharmaceuticals, Inc./Biogen and Roche clinical trials; funding from Famiglie SMA Italy, Italian Telethon and SMA Europe; RSF CHERISH; grants from and advisor fees from Biogen; grants from Cytokinetics; outside the submitted work: advisor for Novartis and Roche; advisory capacity to non-profit organisations: Cure SMA, SMA Europe, SMA Reach (UK) and the Spinal Muscular Atrophy Foundation; data safety monitoring board for the AveXis gene transfer study and Roche Moonfish studies; JK: advisory boards for AveXis, Biogen, Ionis Pharmaceuticals, Inc. and Roche; CAC: advisory boards for AveXis, Biogen, Ionis Pharmaceuticals, Inc. and Roche; grants from Biogen, Ionis Pharmaceuticals, Inc., the National Institutes of Health and the Spinal Muscular Atrophy Foundation; NK: advisory boards for Biogen; consulting employees of and hold stock/stock options in Ionis Pharmaceuticals, Inc.; PS, SG and WF: employees of and hold stock/stock options in Biogen. **Acknowledgments** This study was sponsored by Ionis Pharmaceuticals, Inc. (Carlsbad, CA, USA) and Biogen (Cambridge, MA, USA). Writing and editorial support for the preparation of this poster was provided by Excel Scientific Solutions (Southport, CT, USA): funding was provided by Biogen.

The 22nd International Annual Congress of the World Muscle Society | 3-7 October 2017 | Saint Malo, France